Adenosine signature genes associate with tumor regression in renal cell carcinoma (RCC) patients treated with the adenosine A2A receptor (A2AR) antagonist, CPI-444.

[1]Andrew Hotson, [1]Stephen Willingham, [2]Lawrence Fong, [3]John Powderly II, [4]Jason Luke, [5]Mario Sznol, [6]Saby George, [7]Toni K. Choueiri, [7]Marios Giannakis, [8]Brian Rini, [9]Shivaani Kummar, [10]Erik Evensen, [1]Ian McCaffery, [1]Chunyan Gu, [1]Long Kwei, [1]Ginna Laport, [1]Joe Buggy and [1]Richard Miller

[1]Corvus Pharmaceuticals, Burlingame, CA; [2]University of California, San Francisco, San Francisco, CA; [3]Carolina BioOncology Institute, Huntersville, NC; [4]University of Chicago Medical Center, Chicago, IL, [5]Yale University School of Medicine, New Haven, CT; [6]Roswell Park, Buffalo, NY; [7]Dana-Farber Cancer Institute, Boston, MA; [8]Cleveland Clinic, Cleveland, OH; [9]Stanford University School of Medicine, Stanford, CA; [10]Basis Bioscience, Foster City, CA

Background

- Adenosine blocks T-cell activation and promotes myeloid suppression
- CPI-444 is an oral small molecule antagonist of the adenosine 2A receptor (A2AR) that has shown efficacy in animal models and is associated with T cell activation. [a,b]
- Ongoing clinical trial of CPI-444 +/- anti-PD-L1 atezolizumab demonstrates tumor responses to monotherapy and combination in multiple indications including renal cell carcinoma (RCC). [c,d]
- Future trials in RCC would benefit from a biomarker that predicts patient response.

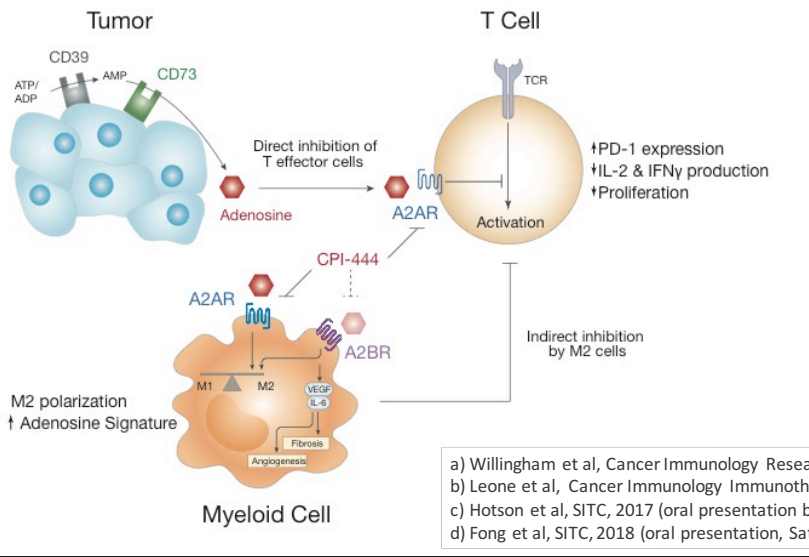
Tumor
T Cell
CD39
ATP/ADP
AMP
CD73
TCR
Direct inhibition of T effector cells
Adenosine
A2AR
Activation
↑PD-1 expression
↑IL-2 & IFNγ production
↑Proliferation
CPI-444
A2AR
A2BR
Indirect inhibition by M2 cells
M1
M2
VEGF
IL-6
Fibrosis
Angiogenesis
M2 polarization
↑ Adenosine Signature
Myeloid Cell

a) Willingham et al, Cancer Immunology Research, 2018
b) Leone et al, Cancer Immunology Immunotherapy, 2018
c) Hotson et al, SITC, 2017 (oral presentation by Luke, J)
d) Fong et al, SITC, 2018 (oral presentation, Sat @ 4:40pm)

Phase 1/1b Clinical Study with CPI-444

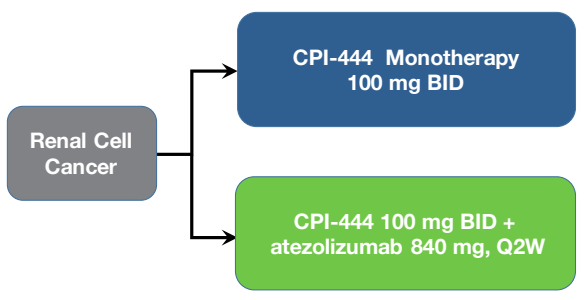
Renal Cell Cancer
CPI-444 Monotherapy 100 mg BID
CPI-444 100 mg BID + atezolizumab 840 mg, Q2W

Eligibility
- Heavily pretreated (median 3 prior therapies)
- Prior anti-PD-(L)1 allowed
- Progressive disease on prior therapy
- No selection for PD-L1 expression

Adenosine Signature: *In vitro* Discovery and *In vivo* Application

Signal Identification

PBMCs → + Adenosine analog (NECA), 48hr →

+TCR Activation

Adenosine Responsive Genes
- Comprised of myeloid cell recruitment and activation
- Inhibition of T cell activation

Identify tumors that express Adenosine Signature (putative adenosine-rich tumor environment)

Applied to Clinical Samples

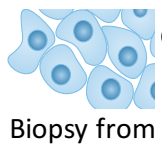

Gene Expression (Nanostring)

Biopsy from patient screening

Adenosine Signature Distribution Across Patients

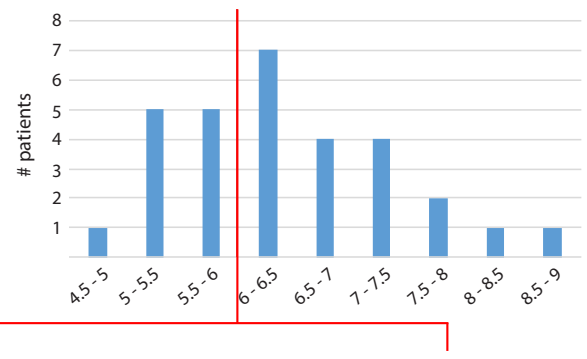
patients
8
7
6
5
4
3
2
1
4.5 - 5
5 - 5.5
5.5 - 6
6 - 6.5
6.5 - 7
7 - 7.5
7.5 - 8
8 - 8.5
8.5 - 9

Adenosine Signature Low
No tumor regression

Adenosine Signature High
Enriched for tumor responders to CPI-444

Adenosine Signature and Co-expressed Genes Identified in Patient Subset by Unsupervised Clustering

A separate patient subset is low for adenosine signature and expresses alternate biological pathways

- Gene expression was collected from pre-treatment biopsies
- Expression was correlated across patients and clustered
- Biology self-organized to reveal modules of gene co-expression

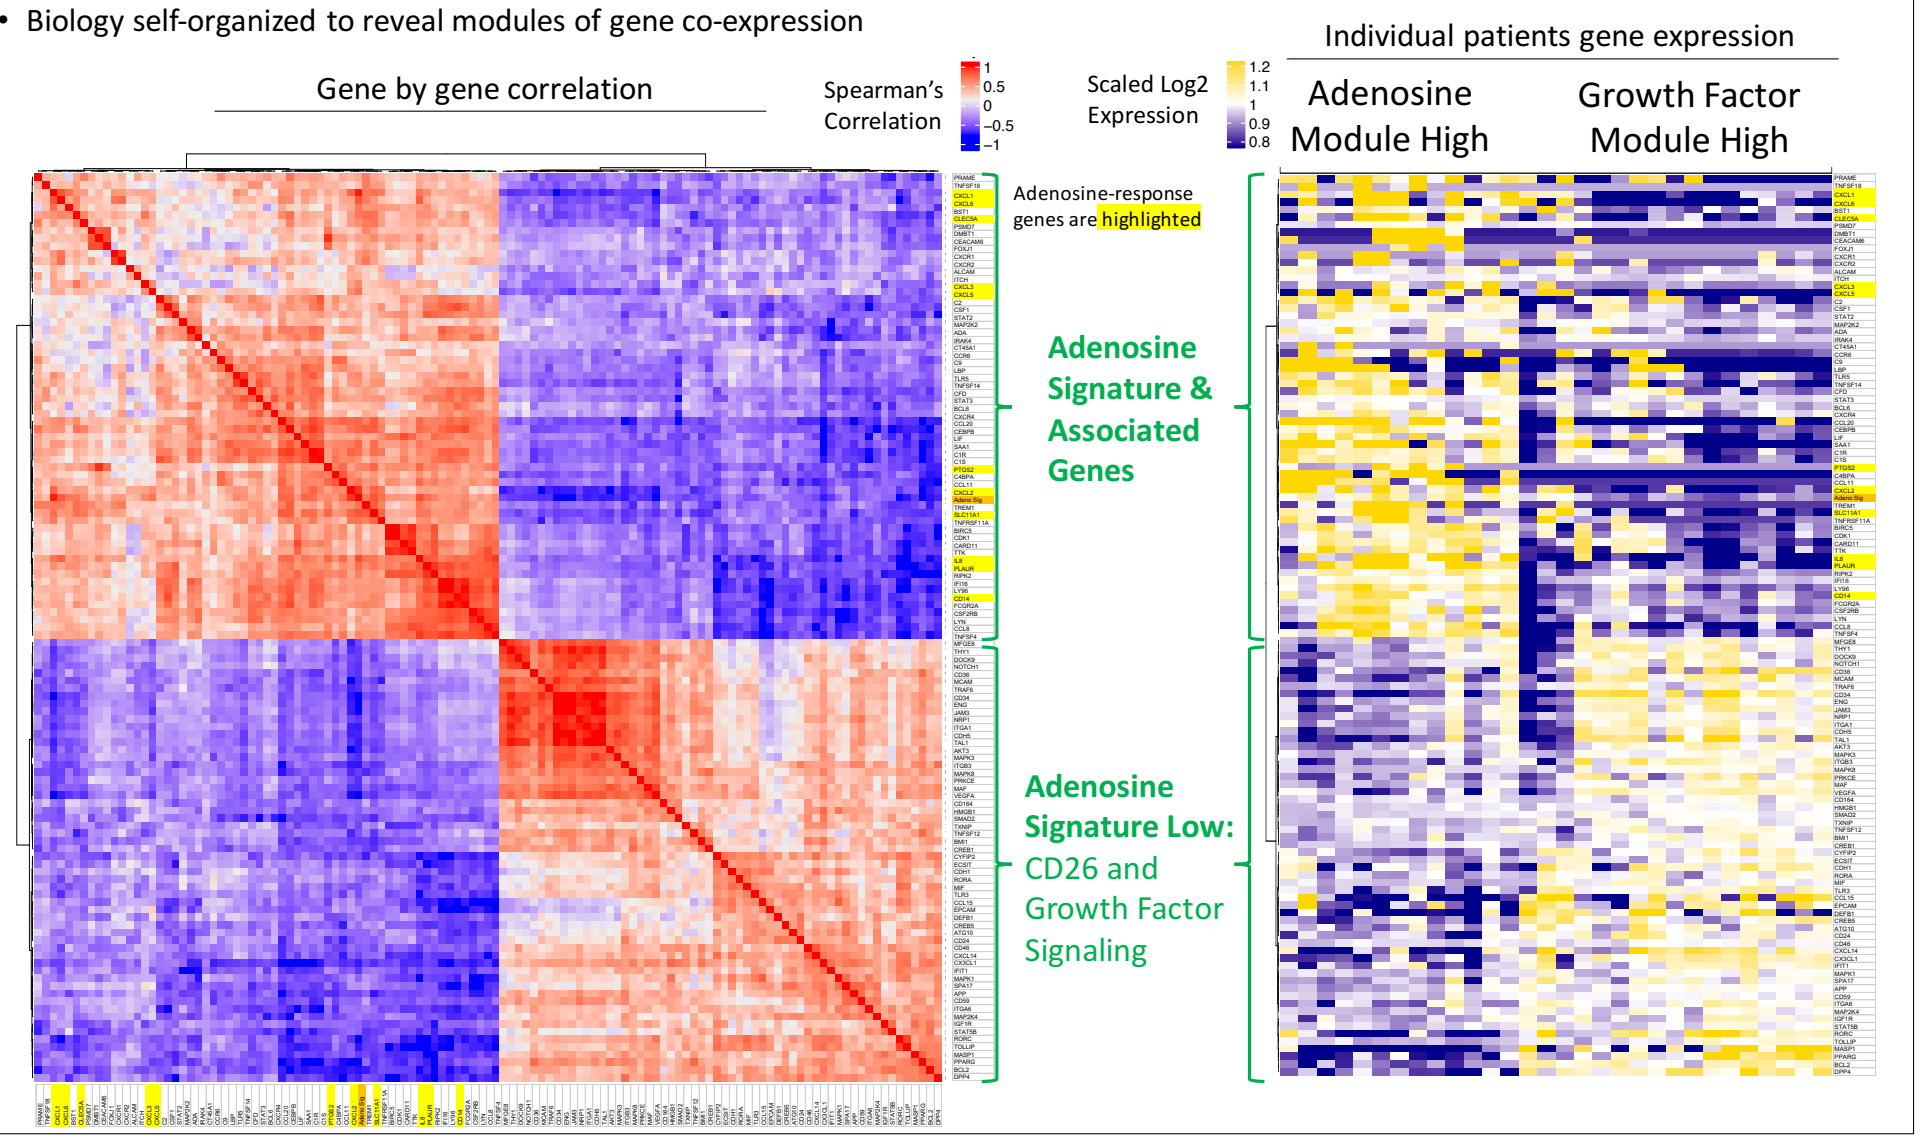
Gene by gene correlation
Spearman's Correlation
1
0.5
0
−0.5
−1
Scaled Log2 Expression
1.2
1.1
1
0.9
0.8
Individual patients gene expression
Adenosine Module High
Growth Factor Module High
Adenosine-response genes are highlighted
Adenosine Signature & Associated Genes
Adenosine Signature Low: CD26 and Growth Factor Signaling

CD26 Negatively Correlates with Adenosine Signature

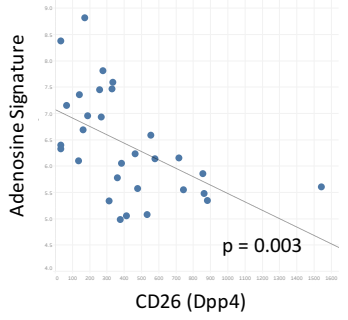
Adenosine Signature
p = 0.003
CD26 (Dpp4)

- CD26 is a binding partner for soluble adenosine deaminase (ADA)
- ADA decreases local adenosine concentration

Adenosine Signature Biomarker and Outcome

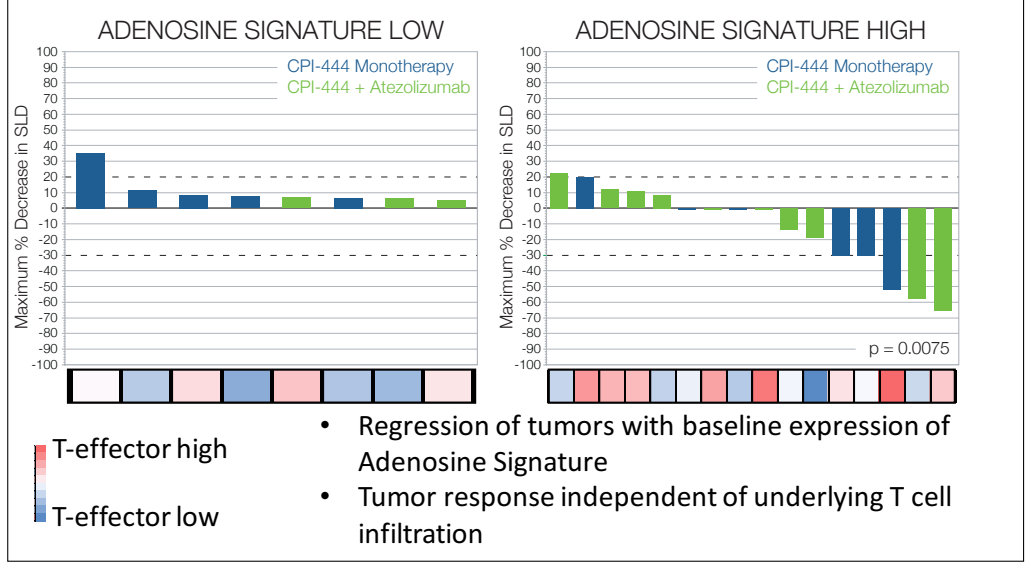
ADENOSINE SIGNATURE LOW
ADENOSINE SIGNATURE HIGH
CPI-444 Monotherapy
CPI-444 + Atezolizumab
Maximum % Decrease in SLD
100
90
80
70
60
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
-100
p = 0.0075
T-effector high
T-effector low

- Regression of tumors with baseline expression of Adenosine Signature
- Tumor response independent of underlying T cell infiltration

Adenosine Signature and Associated Biology

Model of distinct RCC subclasses

Adenosine Microenvironment	Non-Adenosine Microenvironment
• Myeloid recruitment • +/- T cell infiltration • Complement	• Growth factor response • +/- T cell infiltration • Complement inhibition • CD26
Responsive to A2AR inhibition	*Adenosine pathway independent tumor*

Summary

- Adenosine-response genes define an Adenosine Signature biomarker that enriches for patients with tumors that respond to A2AR antagonism by CPI-444
- Gene clustering analysis identified two distinct populations of RCC
 1) Adenosine Signature high / growth factor low
 2) Adenosine Signature low and high for growth factor response genes & CD26
- Enables future studies to employ Adenosine Signature for identification of sub-groups that associate with tumor response